Montana Department of LABOR & INDUSTRY

Workplace Violence in Montana

Using Qualitative Workers' Compensation Data to Examine Trends in Occupational Safety

Andrew Matheison, MPA
Montana Occupational Health and Safety Surveillance Program (MOHSS)
Montana Department of Labor & Industry
(406) 444-4696 | Andrew.Matheison2@mt.gov

1 PURPOSE

Acts of violence in the workplace present a serious hazard to the physical and mental health of workers. High-profile incidents of violence have prompted government and industry stakeholders to assess prevalence and develop response strategies. In Montana, a desire to formulate solutions grounded in data prompted the MOHSS Program to identify incidents of workplace violence reported to the Montana Workers' Compensation Administration Network, administered by the Montana Department of Labor & Industry.

OBJECTIVES:

- Determine the number of reported incidents of workplace violence in Montana
- Examine trends in workplace violence within industries and occupations
- Examine narrative details on instigators of workplace violence

2 BACKGROUND

Workplace violence has been a known occupational hazard and public safety concern for decades. Publications on the prevalence, specifics, and prevention of workplace violence have been released by the Federal Bureau of Investigation, the Occupational Safety and Health Administration, the Bureau of Labor Statistics, and the National Institute for Occupational Safety and Health. Much of this research focuses on fatal occupational injuries from acts of violence, but the definition of violence is much broader, including non-fatal assaults as well as harassment and intimidation. Non-fatal incidents of workplace violence have received far less attention, presenting a potentially under-mitigated threat to workers.

3 METHODS

Most workers' compensation claims in Montana contain an accident description, a short narrative completed by the worker or a representative describing the accident which led to the injury. Claims filed from 2019 to 2024 were reviewed to determine if they had an accident description conclusively pointing to workplace violence. For a claim to be accepted as workplace violence, it needed to identify another person as acting in a capacity to cause harm to the injured worker. Claims identified as having been caused by acts of workplace violence were examined to determine industry and occupational factors as well as relational trends between the injured worker and the individual instigating the workplace violence.

Reported Injuries from Workplace Violence

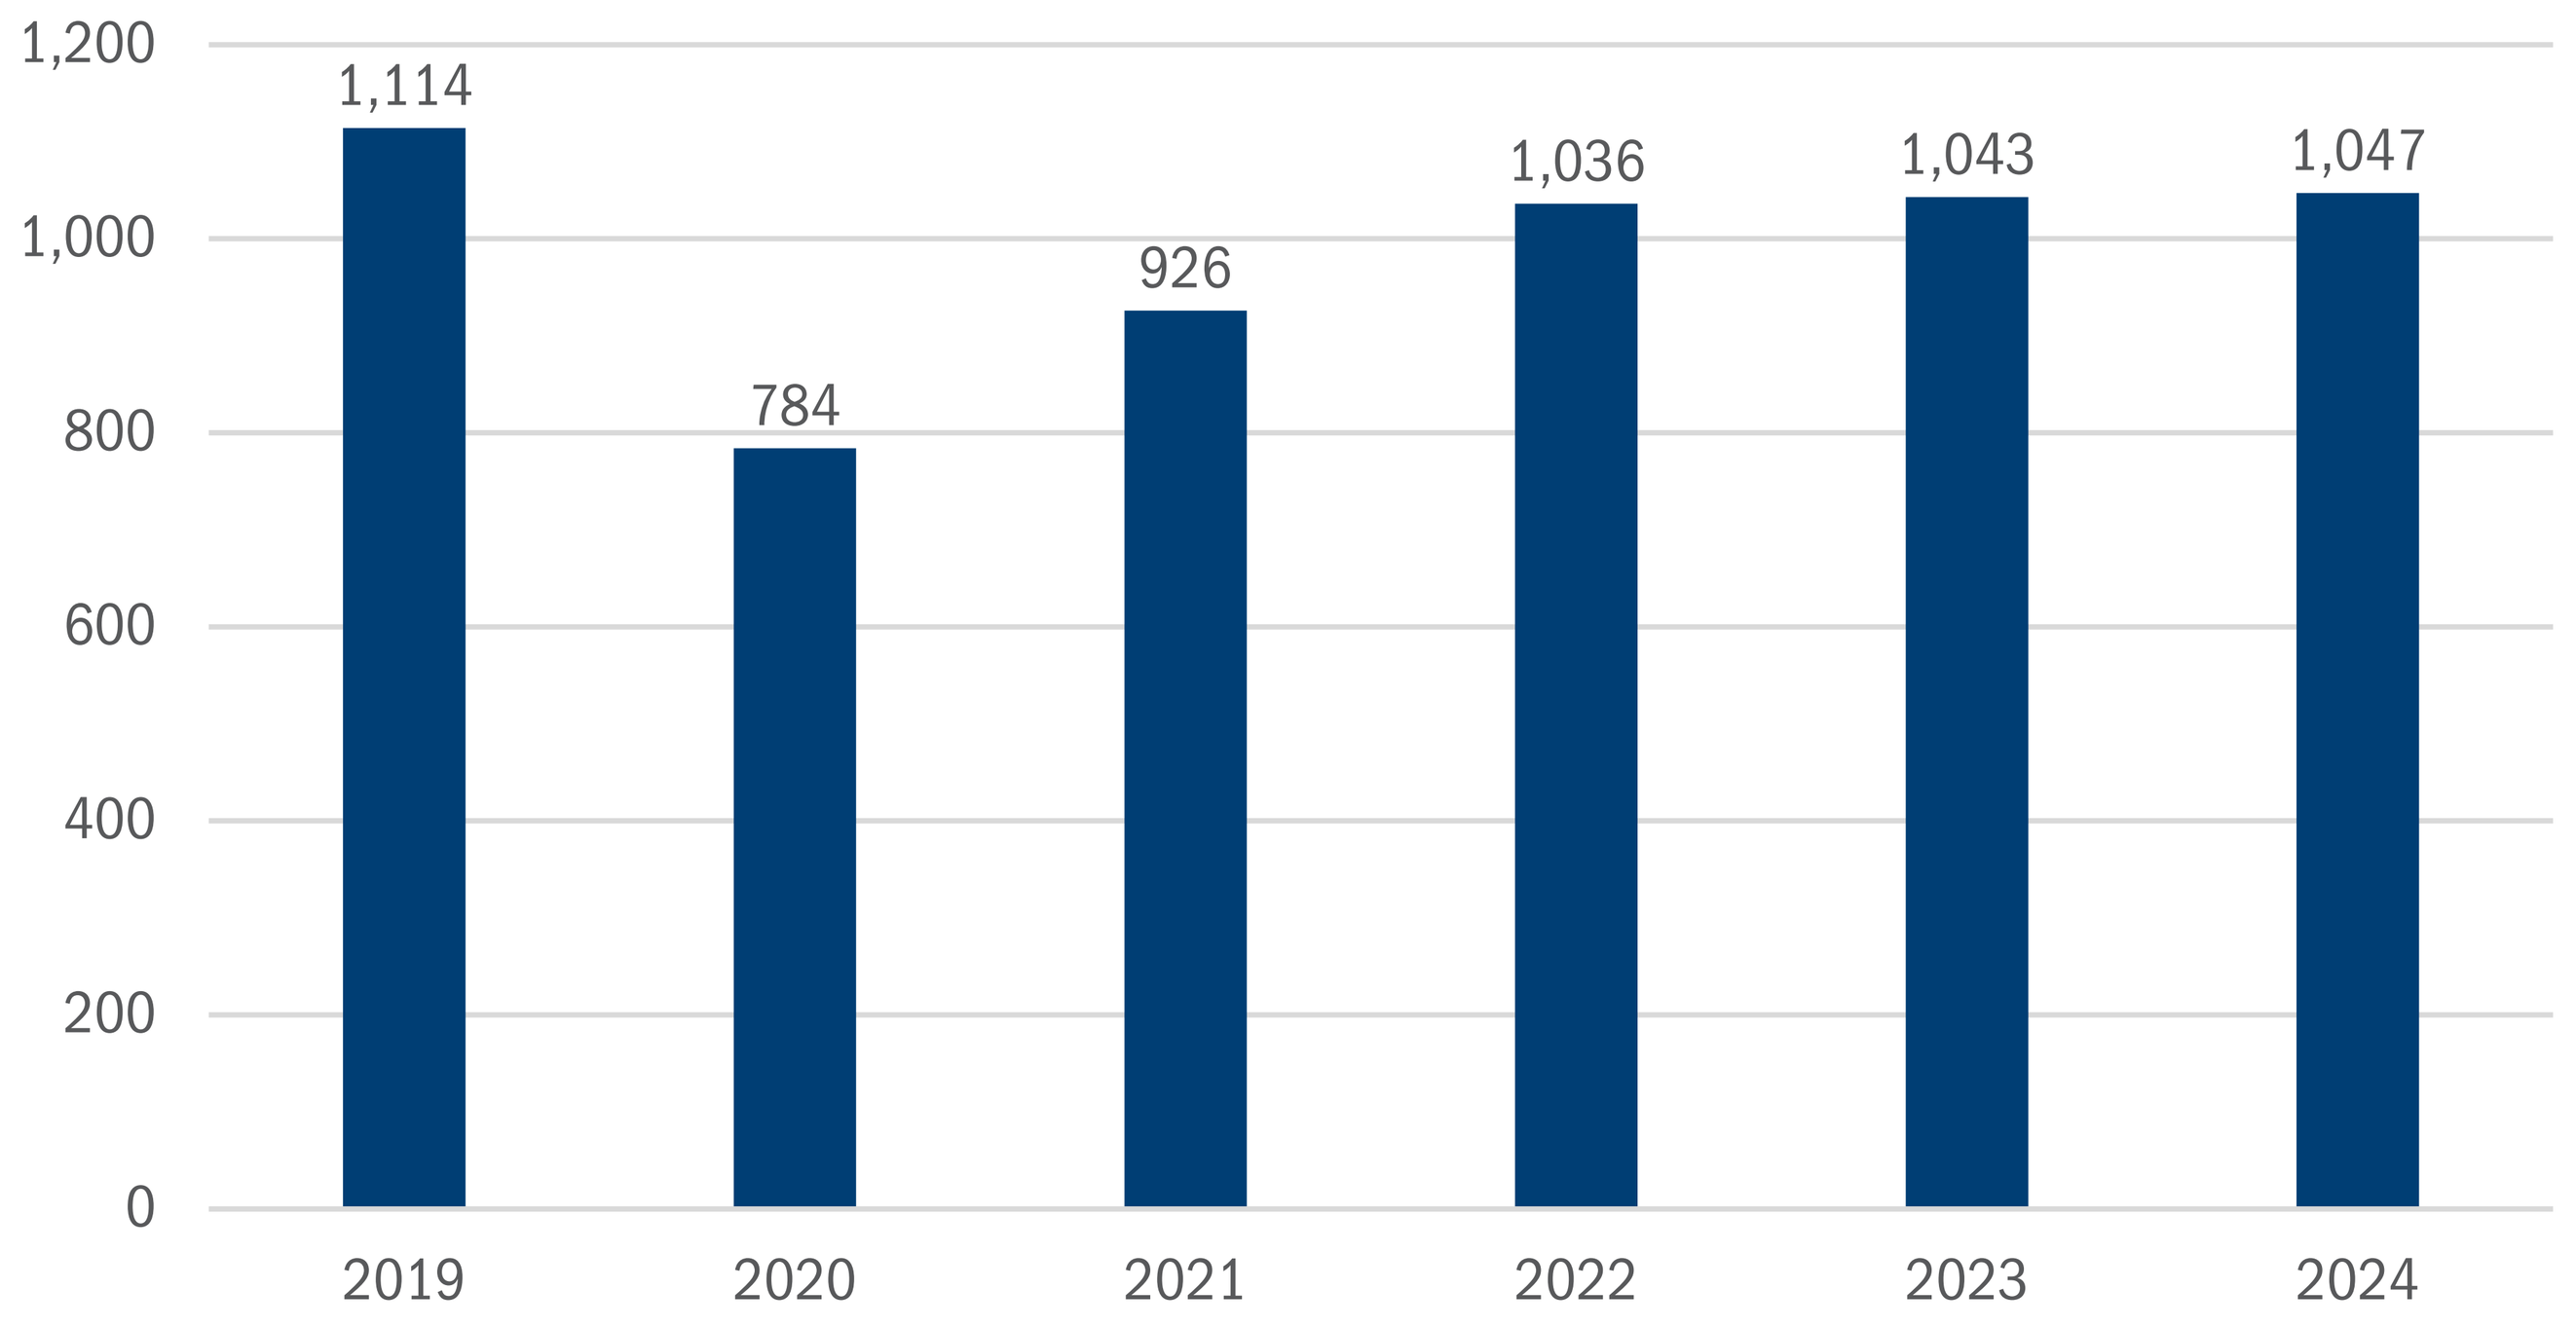
1,200
1,000
800
600
400
200
0
1,114
784
926
1,036
1,043
1,047
2019
2020
2021
2022
2023
2024

Actual vs Relative Incidence of Reported Workplace Violence

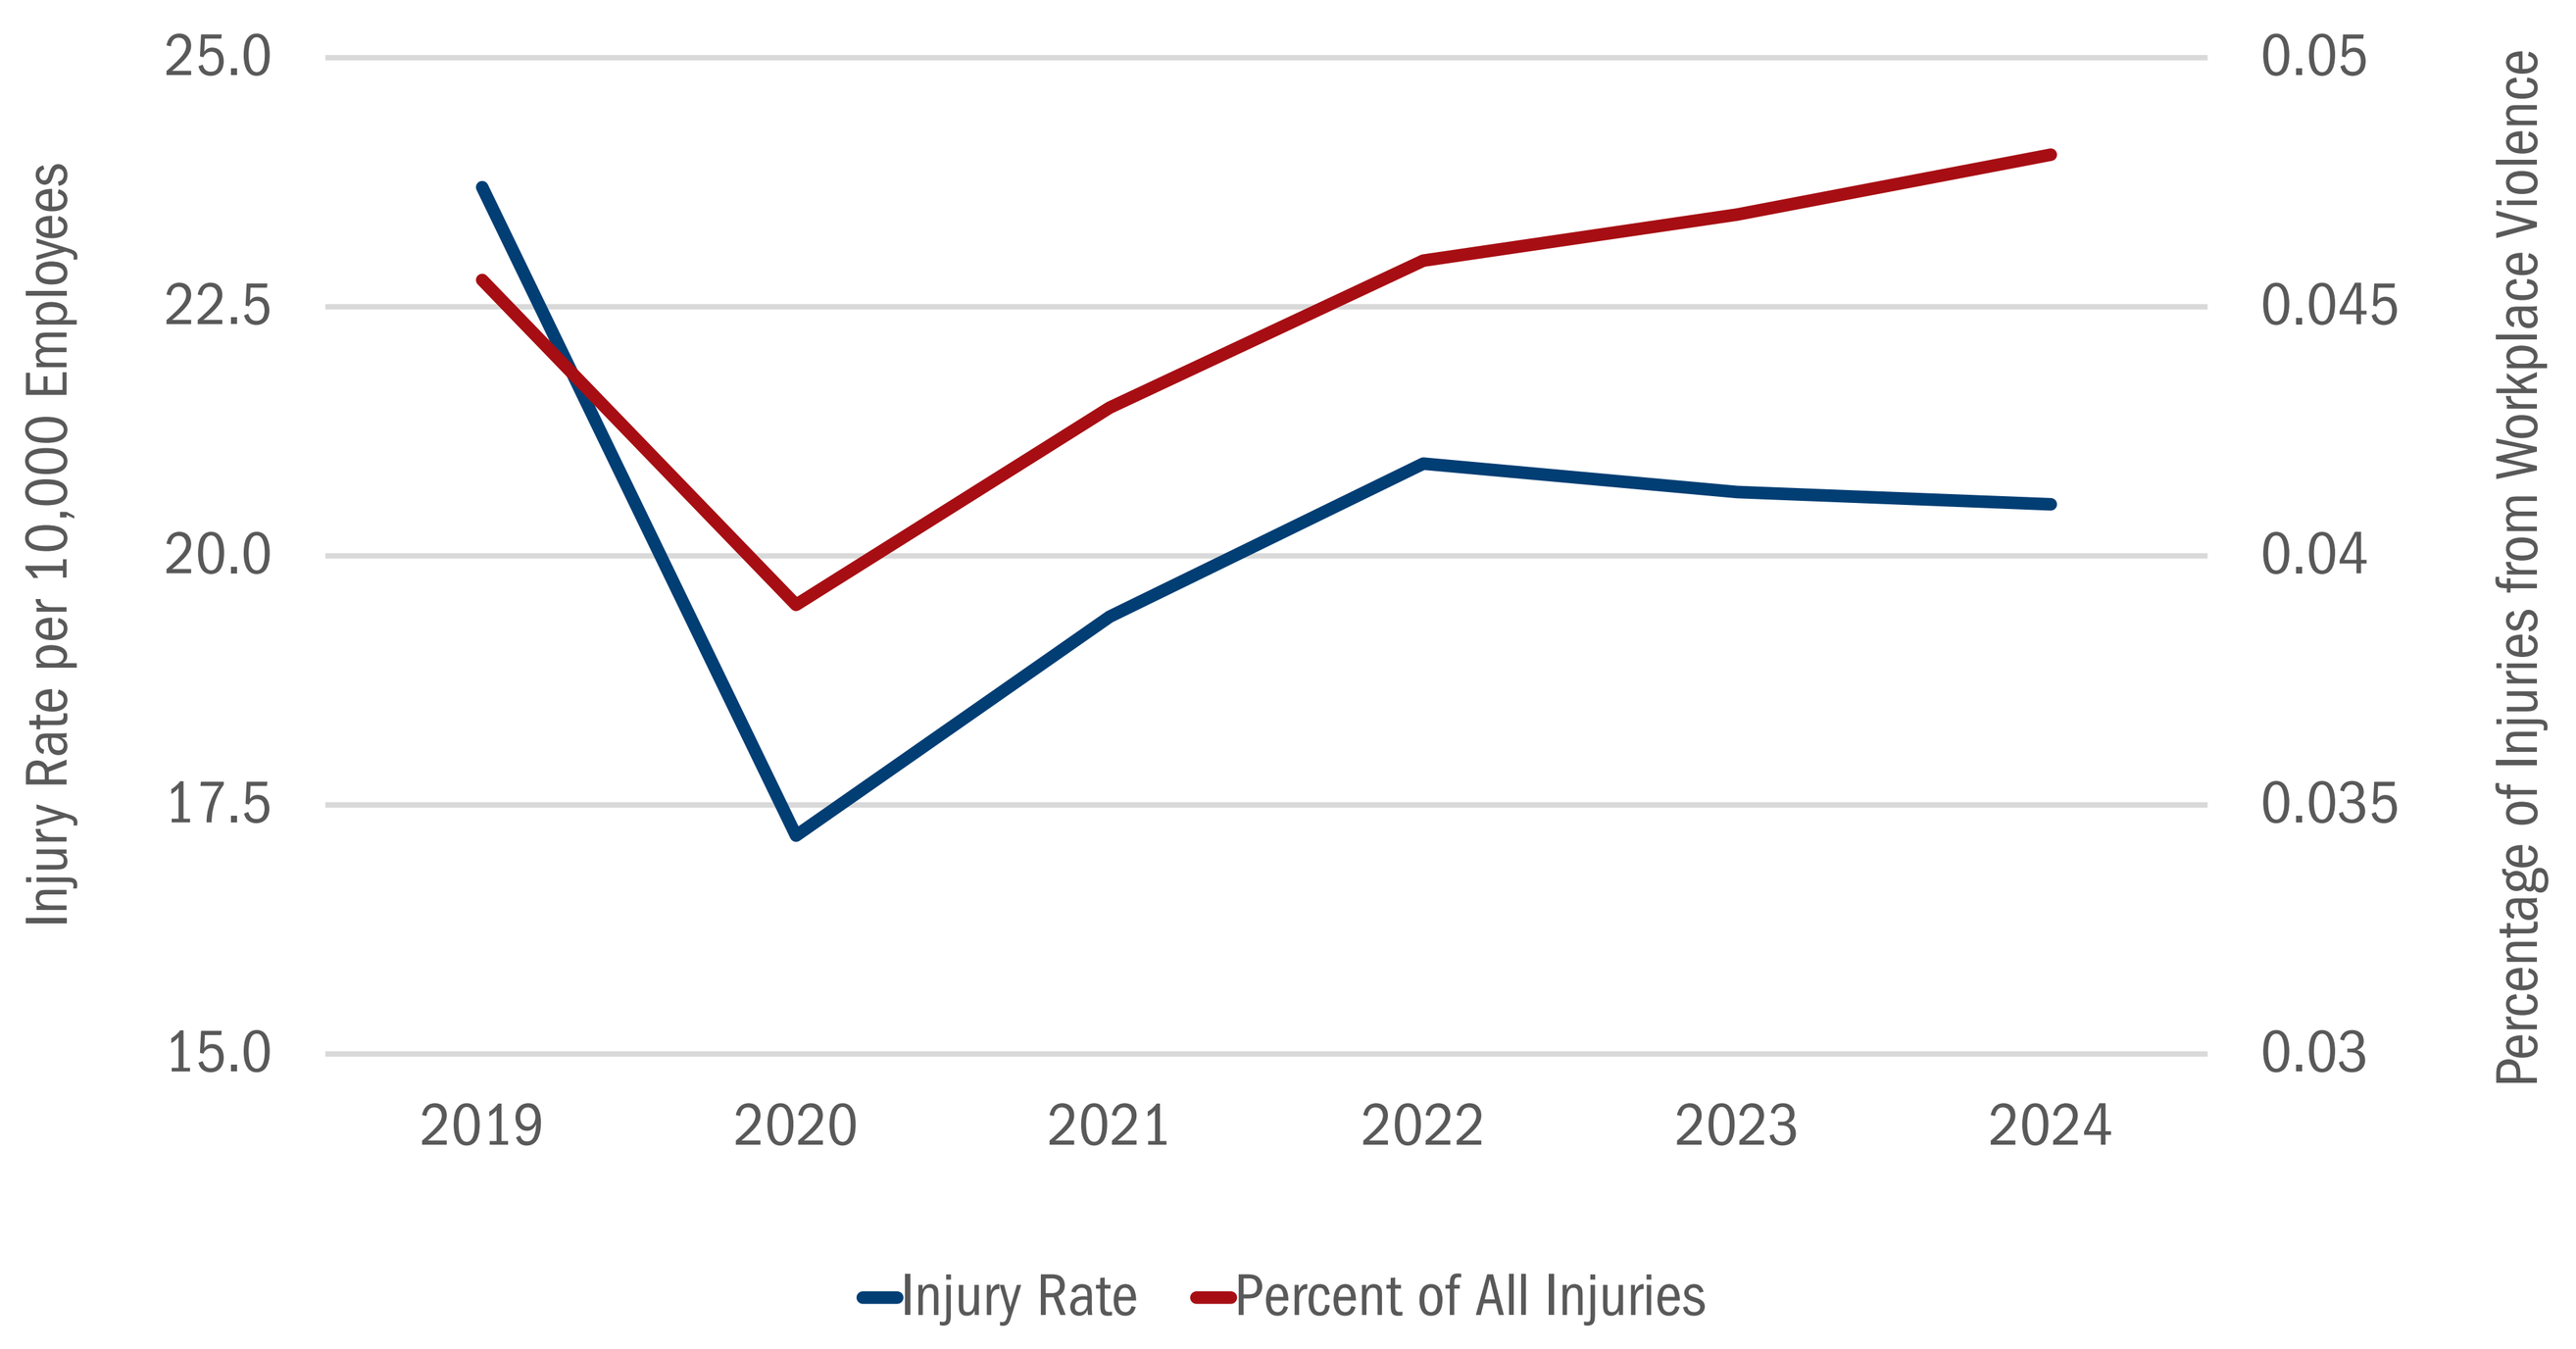
Injury Rate per 10,000 Employees
25.0
22.5
20.0
17.5
15.0
0.05
0.045
0.04
0.035
0.03
Percentage of Injuries from Workplace Violence
2019
2020
2021
2022
2023
2024
Injury Rate
Percent of All Injuries

Rate of Injury from Acts of Workplace Violence per 10,000 Employees, Industries with <10 Injuries per 10,000 Employees

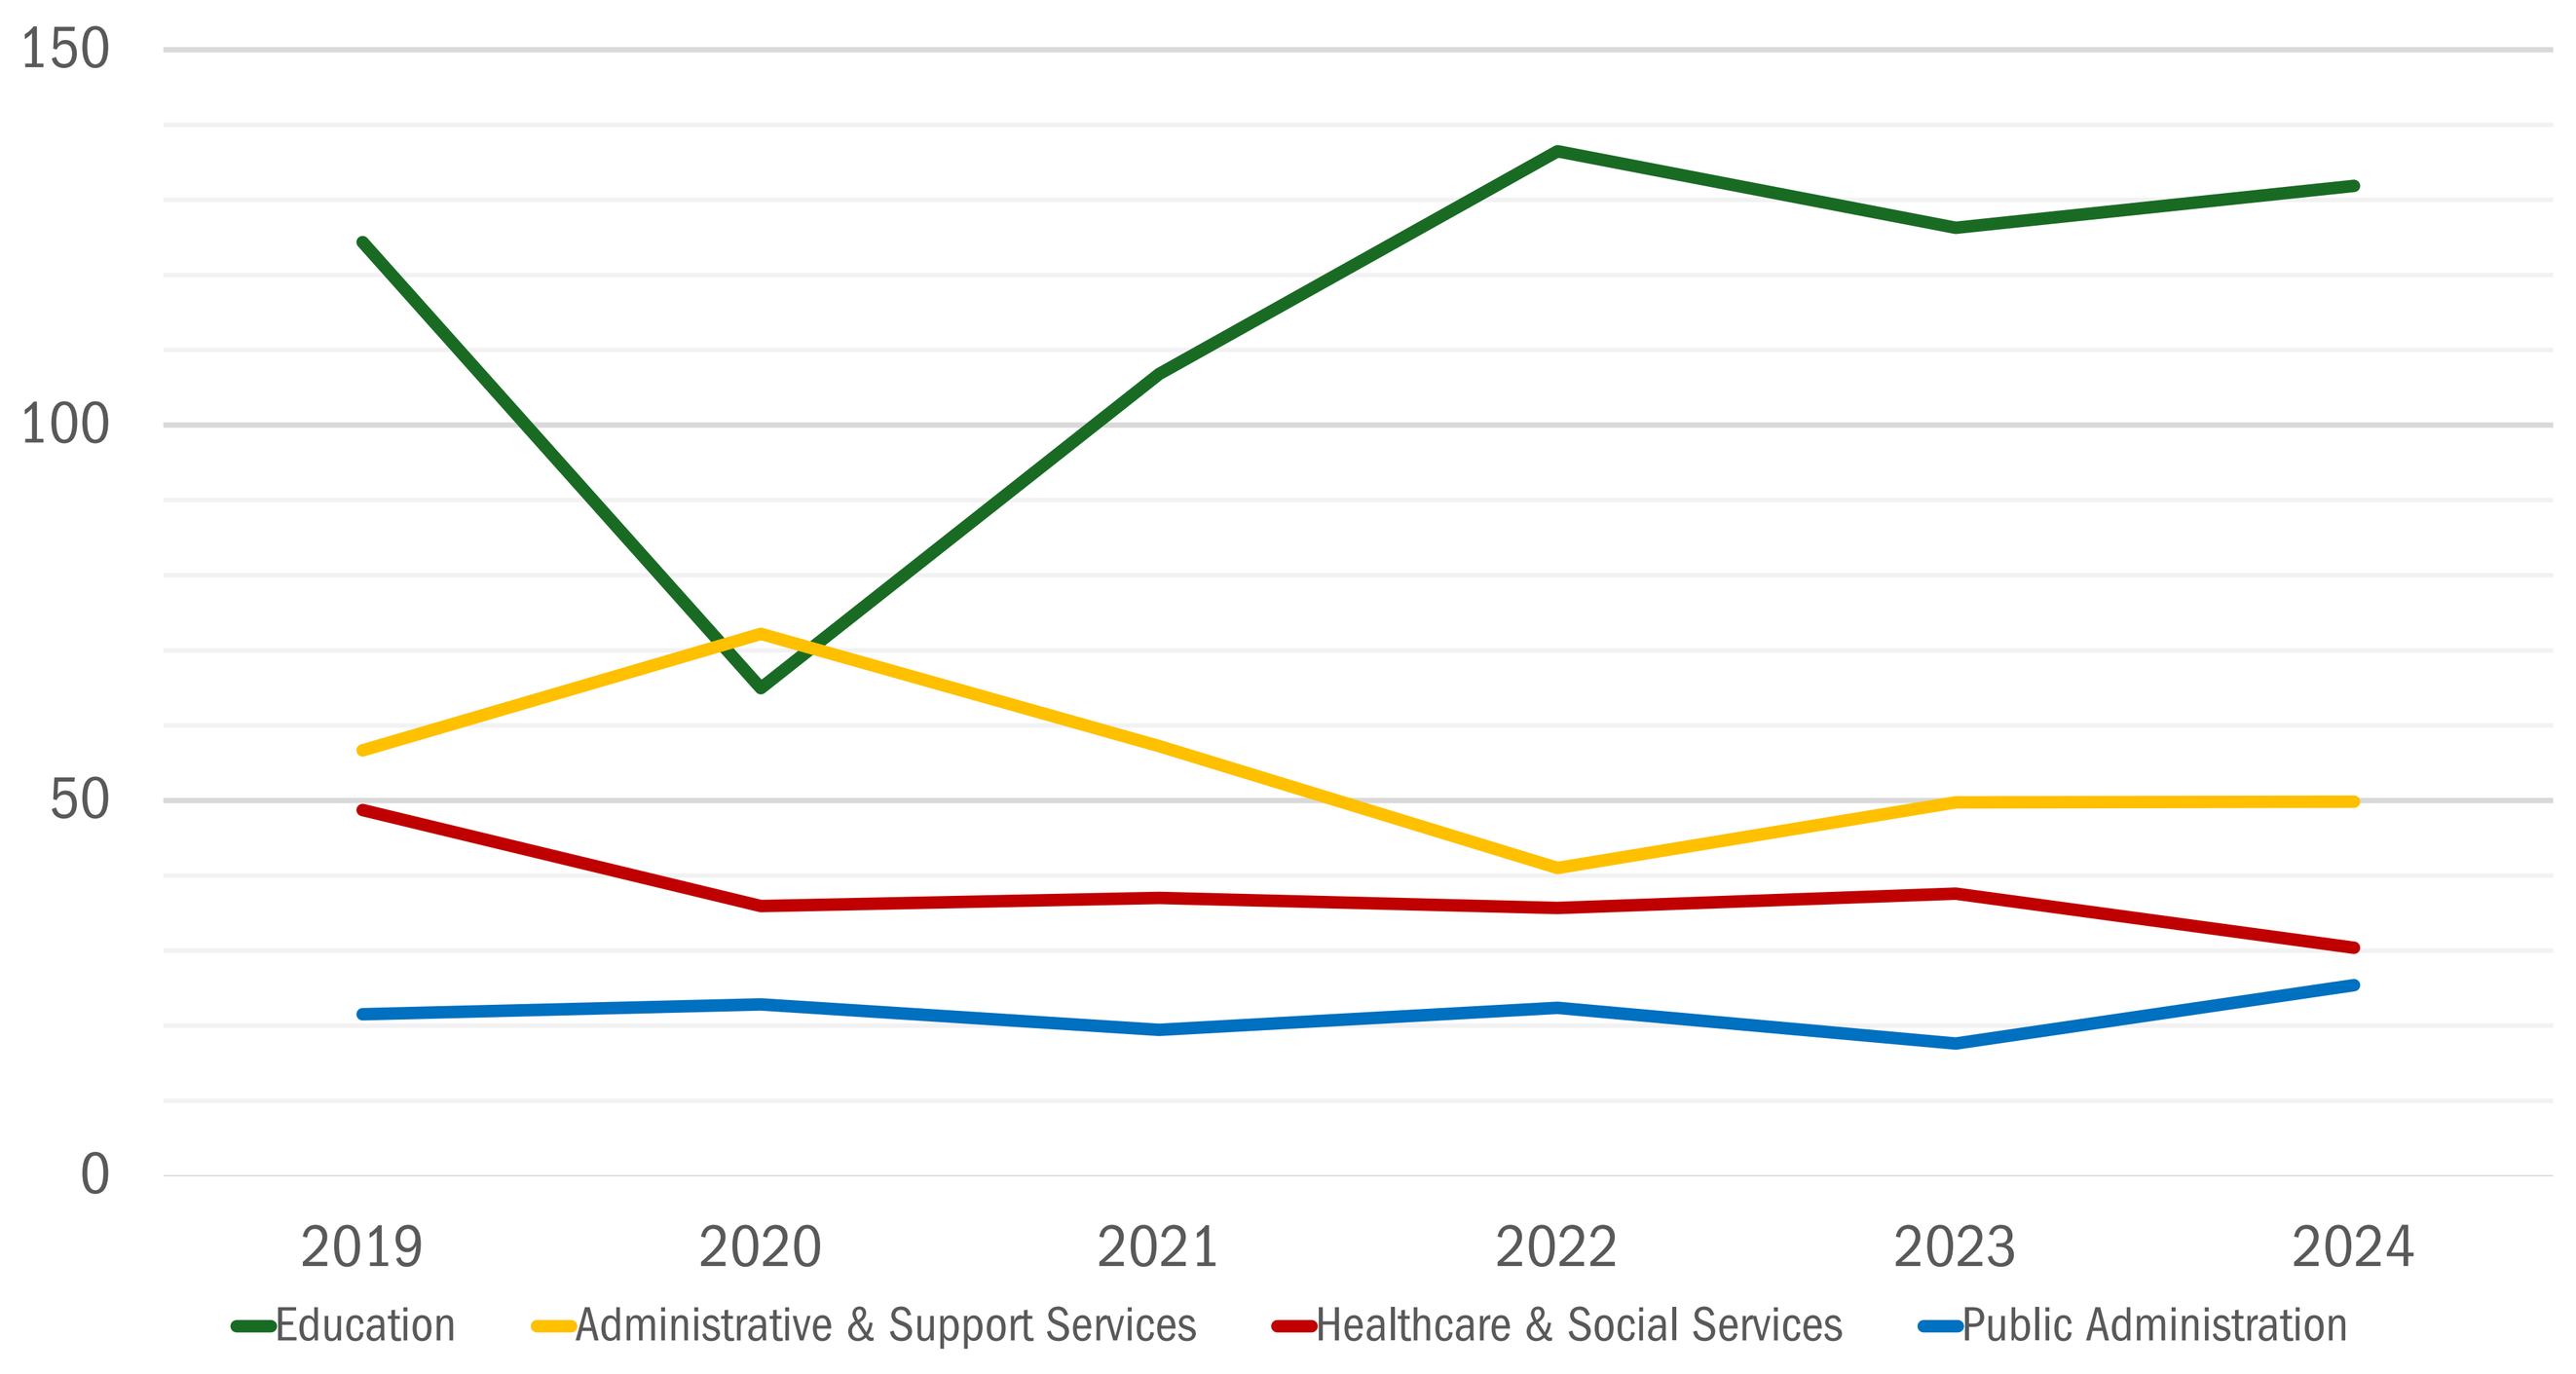
150
100
50
0
2019
2020
2021
2022
2023
2024
Education
Administrative & Support Services
Healthcare & Social Services
Public Administration

Percent of All Report Injuries Attributed to Workplace Violence, Select Industries

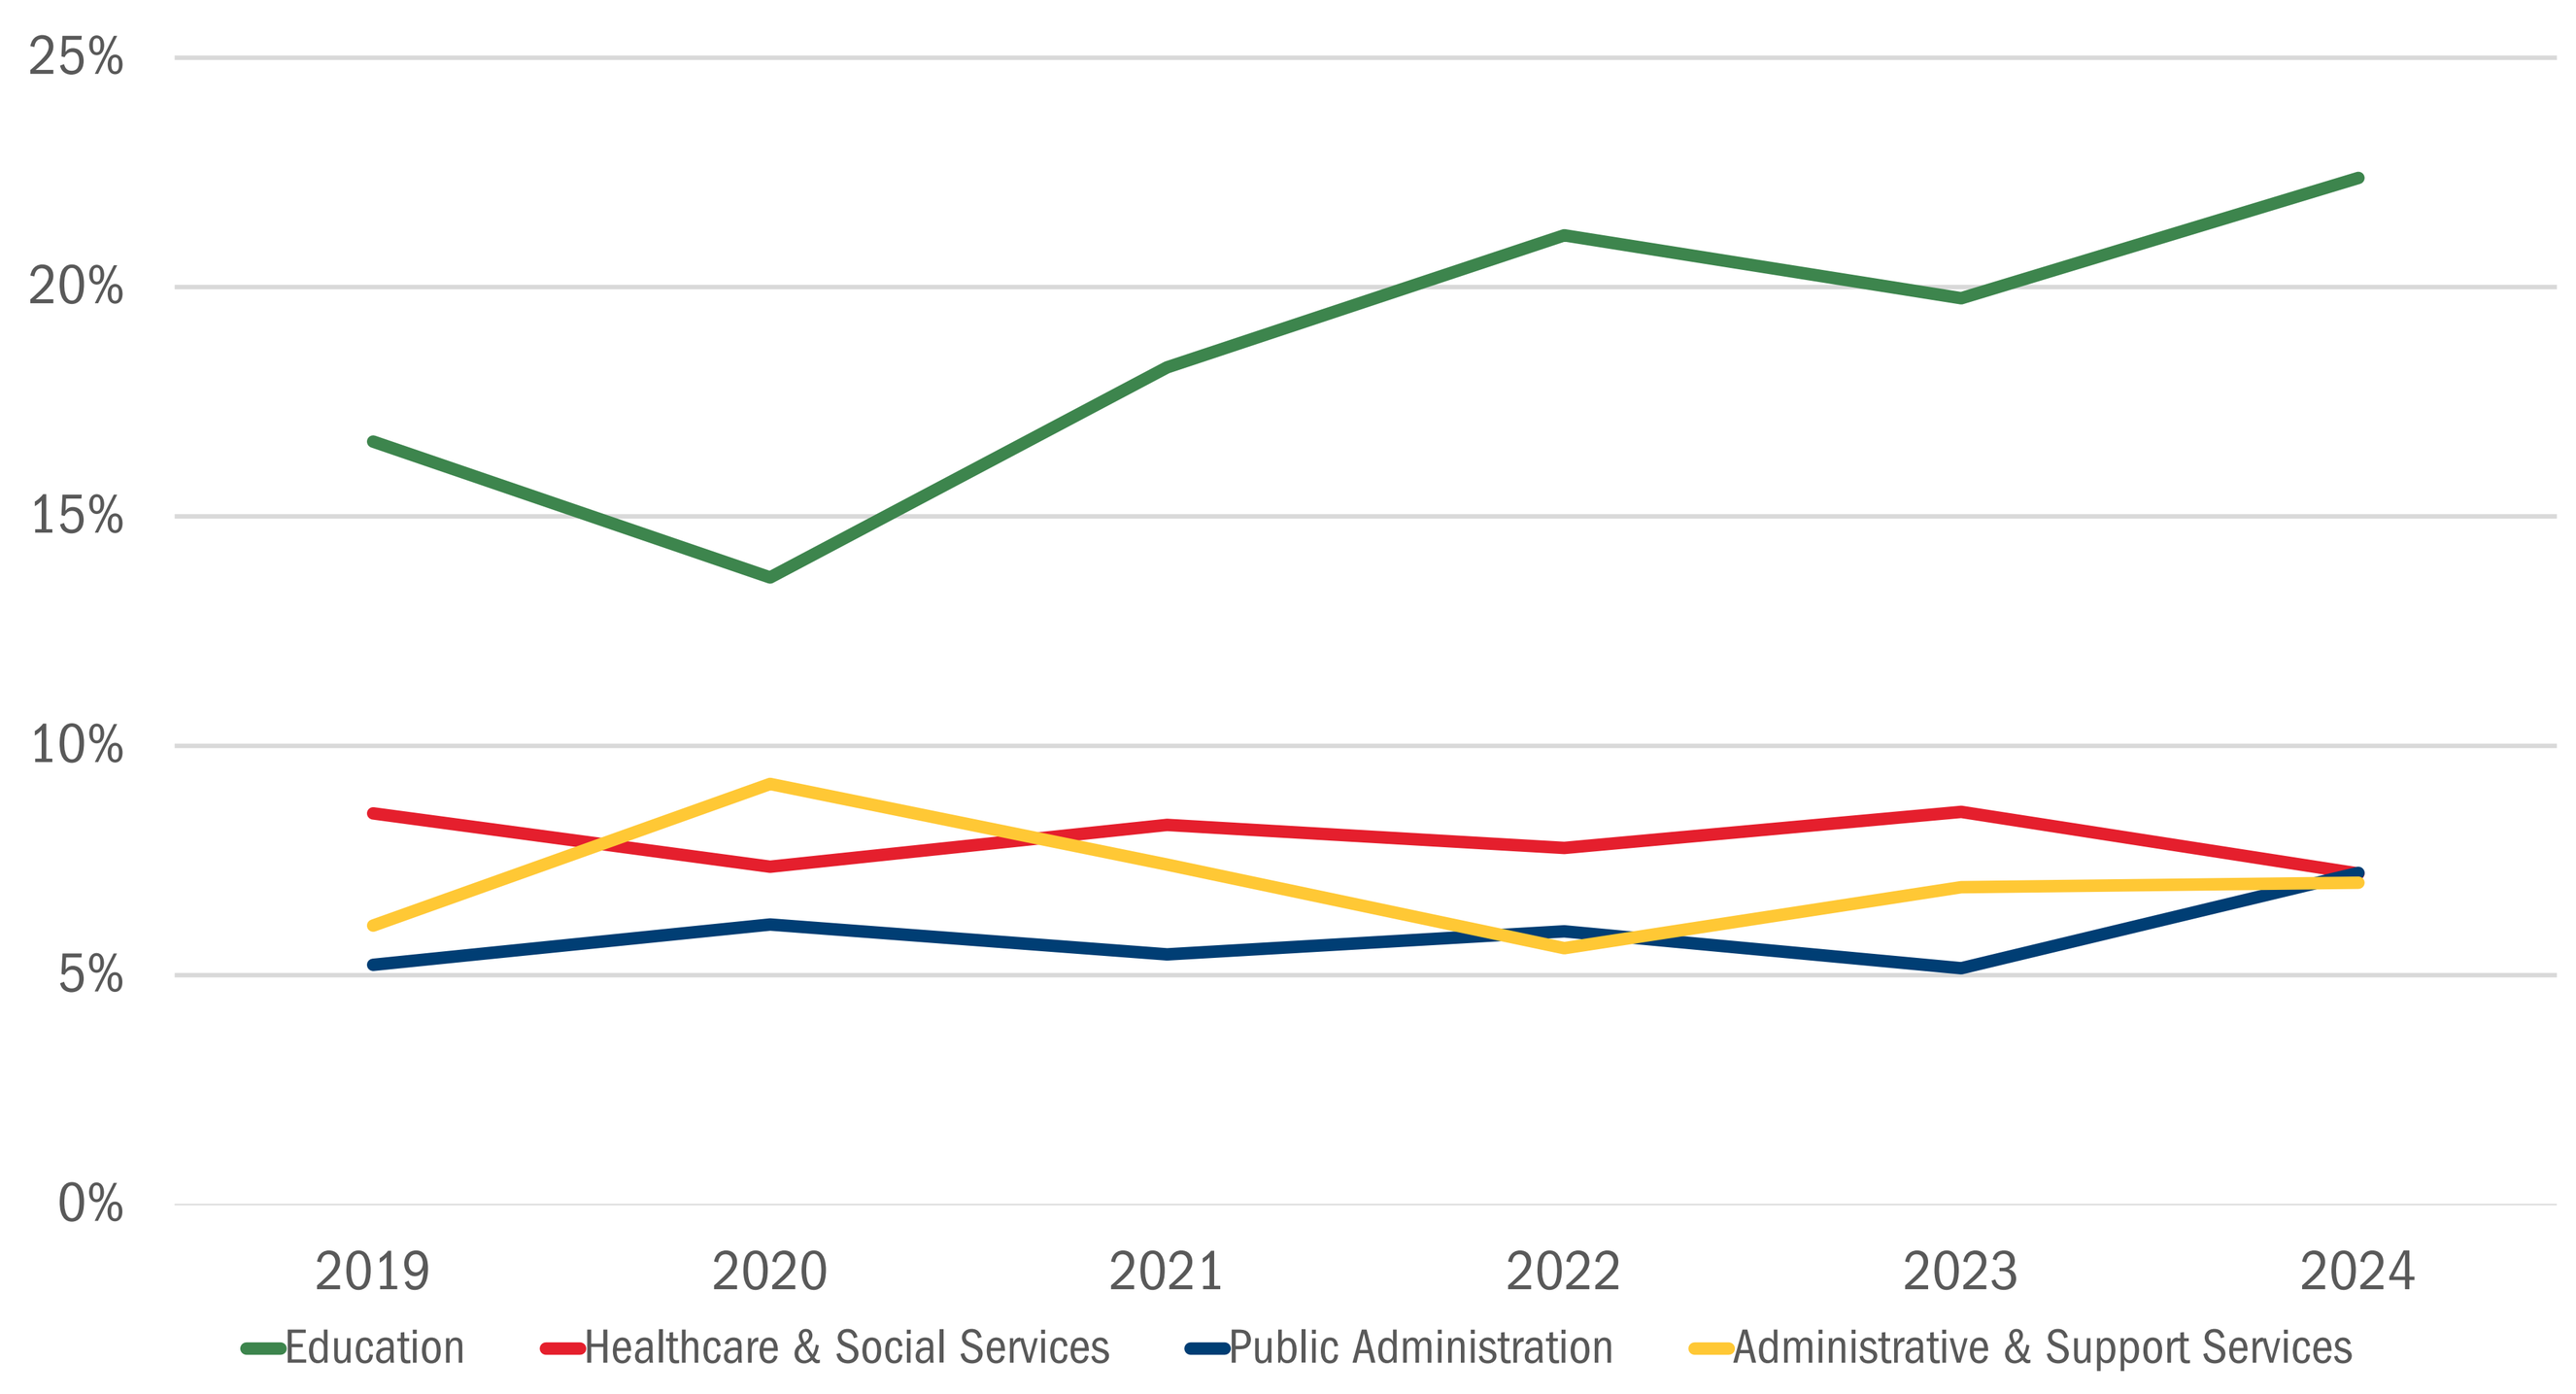
25%
20%
15%
10%
5%
0%
2019
2020
2021
2022
2023
2024
Education
Healthcare & Social Services
Public Administration
Administrative & Support Services

Reported Injuries from Workplace Violence by Select Occupation of Worker

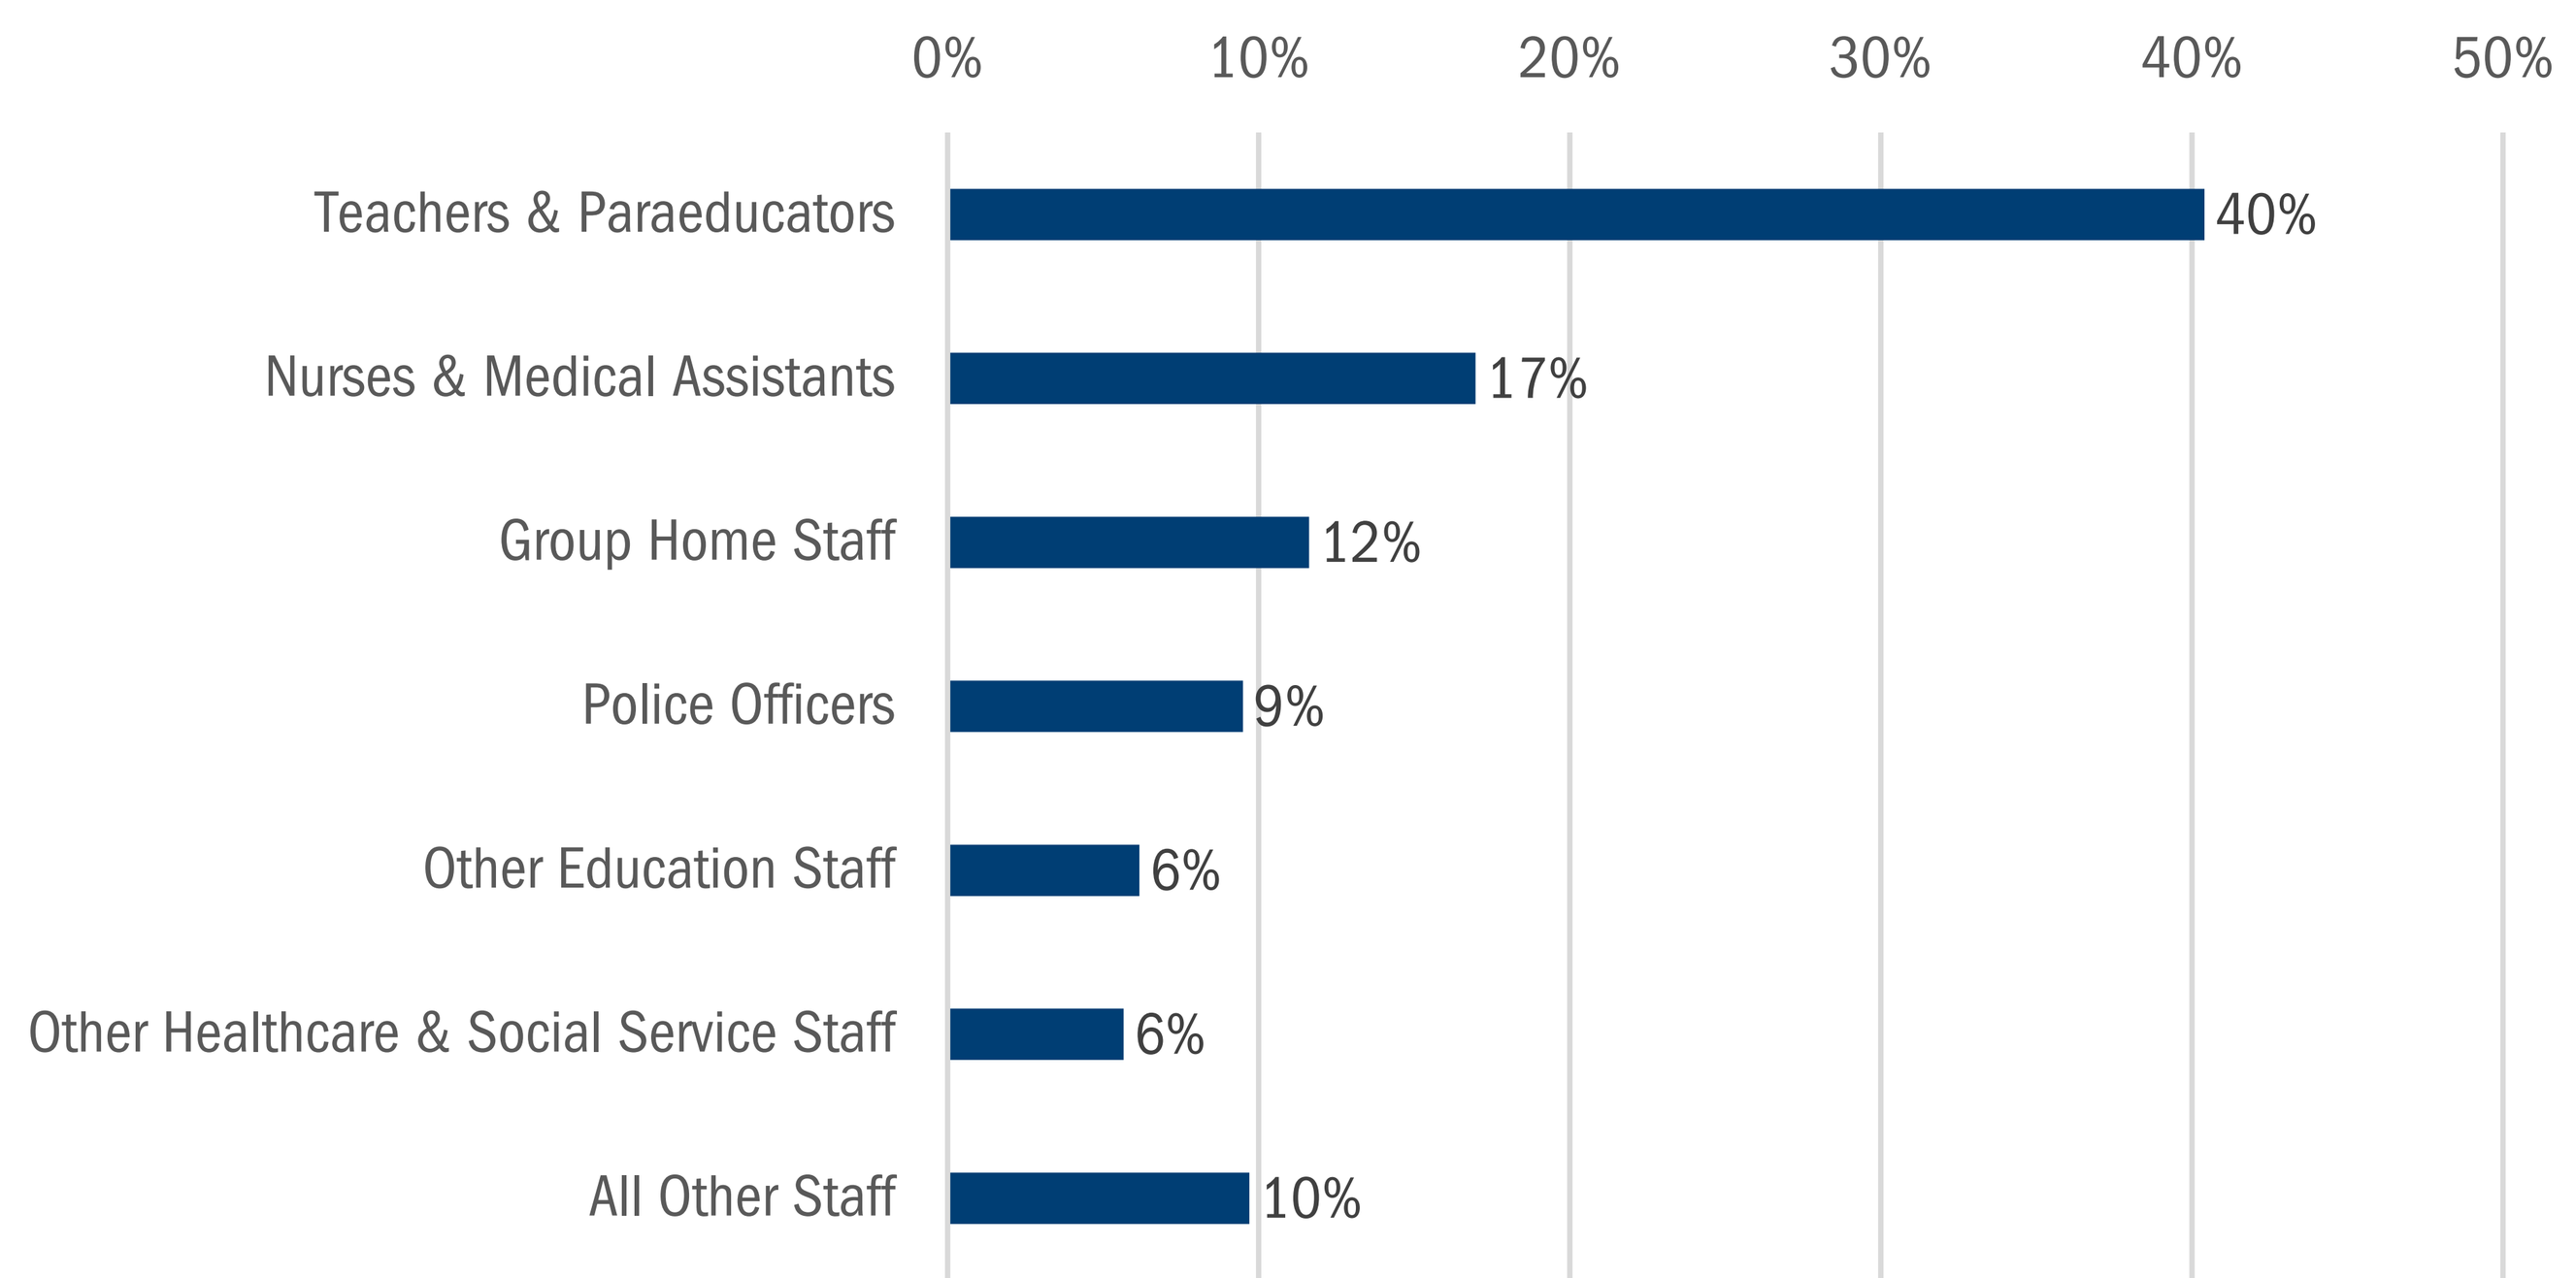
0%
10%
20%
30%
40%
50%
Teachers & Paraeducators
40%
Nurses & Medical Assistants
17%
Group Home Staff
12%
Police Officers
9%
Other Education Staff
6%
Other Healthcare & Social Service Staff
6%
All Other Staff
10%

Instigators of Reported Workplace Violence

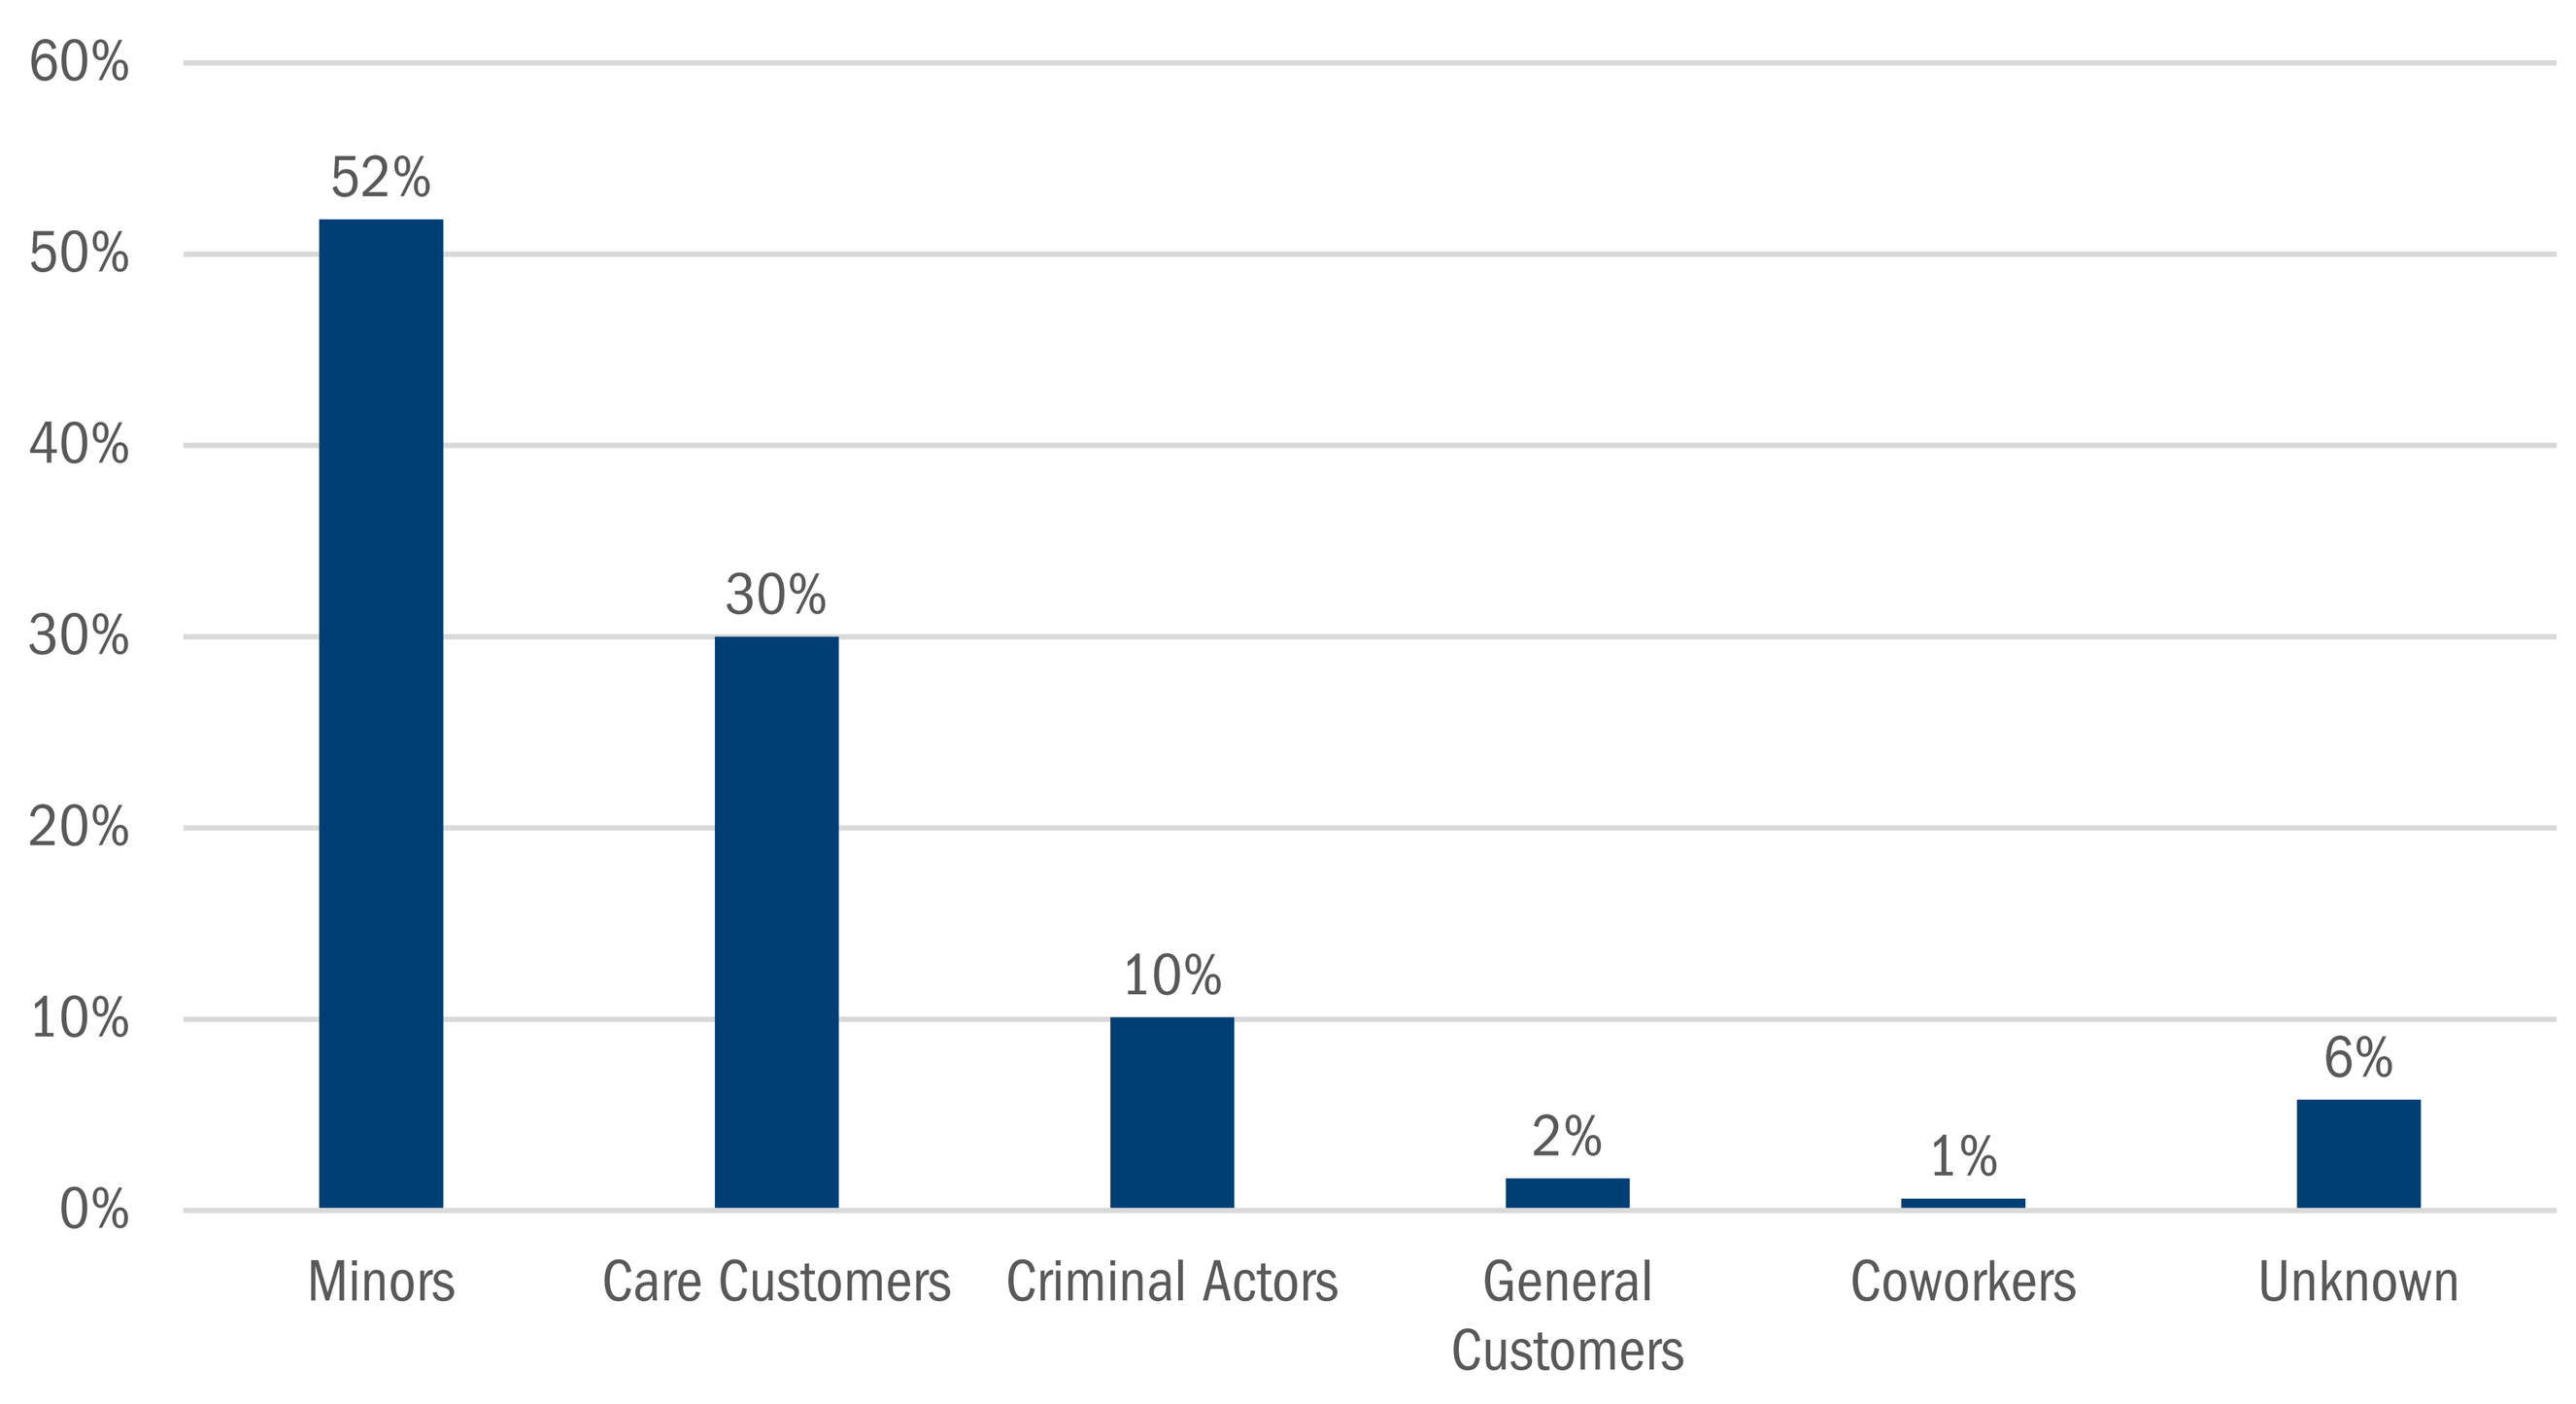
60%
50%
40%
30%
20%
10%
0%
52%
30%
10%
2%
1%
6%
Minors
Care Customers
Criminal Actors
General Customers
Coworkers
Unknown

4 DISCUSSION & NEXT STEPS

This research found:

- The count of reported workplace violence incidents remained steady from 2019 to 2024. The per capita rate of workplace violence incidents declined as employment grew, while the percentage of workplace injuries attributed to workplace violence increased as other sources of workplace injury declined.
- Reported workplace violence incidents were heavily concentrated in several industries and occupations, with the highest rates reported by paraeducators, teachers, nurses, social workers, and police officers.
- Over 80% of reported workplace violence incidents involved child or adult care customers (such as students or patients) aggressing on workers, with only a small percentage of incidents involving conflicts between individuals engaging in or incarcerated for criminal acts, adult customers outside of a caretaker relationship, or violence between coworkers.

Additional research is required to overcome substantial limitations, including:

- The defined scope of workers' compensation, which excludes assaults or behaviors that do not lead to injury. This is defined by statute and unlikely to change.
- Underreporting or inadequate reporting of workplace injuries. Literature on workplace injury has posited substantial underreporting of legitimate workplace injuries. Even when injuries are reported, coding errors or detail omissions can prevent researchers from utilizing data.

Recommended next steps would include:

- Addition of significance testing to research.
- Continued annual surveillance to maintain trends and public awareness.
- Surveying of industries and occupations identified as high-risk to validate findings further.
- Development of guidance for high-risk industries and occupations on preventing and responding to incidents of violence.

5 DATA SOURCES

Montana Department of Labor & Industry, Workers Compensation Administration Network (WCAN), Figures 1-6
U.S. Bureau of Labor Statistics, Quarterly Census of Employment and Wages (QCEW), Figures 2-3

ACKNOWLEDGEMENTS

The Montana Occupational Health and Safety Surveillance Program is funded through a grant from the U.S. Department of Health, Centers for Disease Control and Prevention, National Institute for Occupational Safety and Health.